Improvement of Pityriasis Rubra Pilaris Treated With Efalizumab: A Case Report

2674

Stephen Schleicher, MD

DermDx Centers for Dermatology, Hazleton, Pennsylvania, USA

INTRODUCTION

Pityriasis rubra pilaris (PRP) is a rare, chronic skin disorder of unknown etiology. Most cases are sporadic and acquired, although a familial form exists that is transmitted by an autosomal dominant or autosomal recessive mechanism.[1,2] PRP occurs equally in men and women, with a reported incidence of 1 case in 3500 to 5000 patients presenting at dermatologic clinics. PRP exhibits a bimodal age distribution, with peaks in the first and fifth to sixth decades.[3,4]

PRP is characterized initially by small follicular papules that coalesce into yellowish pink scaly plaques, palmoplantar keratoderma, diffuse furfuraceous scale of the scalp, and frequent progression to exfoliative erythroderma. Features distinguishing PRP from other skin conditions include “islands” of spared skin (so-called islands of sparing) within generalized erythroderma, follicular keratotic plugs, and an orange hue of the involved skin.[5] In adults, PRP typically starts on the face and scalp and spreads in the caudal direction. In children, PRP typically begins on the lower half of the body.[6] Palms and soles are almost always involved, and the lesions may expand and coalesce to cover the entire body.

Griffiths divided PRP into 5 categories based on clinical features, age of onset, and prognosis: classic adult type, atypical adult type, classic juvenile type, circumscribed juvenile type, and atypical juvenile type.[4] More recently, an HIV-associated type was added to this classification system.[7] The classic adult type (type I) accounts for 50% of cases and has the best prognosis, with 80% clearing by 3 years, whereas the familial form of the disease may persist throughout life.

Treatment for PRP often includes systemic retinoids (eg, acitretin) as first-line therapy, although the results are variable.[8] Other agents reported to have produced successful results include topical vitamin D analogues, topical keratolytics, methotrexate, cyclosporine, azathioprine, fumaric acid esters, phototherapy, and infliximab.[5,8-12]

Efalizumab, a recombinant humanized monoclonal IgG1 antibody against the α subunit of lymphocyte function–associated antigen (CD11a), is a T-cell modulator.

Efalizumab is approved for the treatment of moderate to severe chronic plaque psoriasis. Although this is its only approved indication, there have been case reports showing efficacy of efalizumab in the treatment of recalcitrant dermatomyositis, severe atopic dermatitis, alopecia areata, and oral erosive lichen planus.[13-16] Two cases of successful efalizumab treatment of patients with PRP have been reported.[17,18] In contrast, the case of a 60-year-old woman who experienced exacerbation of PRP with efalizumab therapy has been reported,[19] confounding the role of efalizumab for treatment of this disease. Here we present the case of a patient with extensive PRP who was successfully treated with a short course of efalizumab therapy.

CASE REPORT

Presentation and History

Medical History

- RB is a 59-year-old man with an 8-year history of PRP involving his trunk, arms, legs, and hands.

Treatment History

- The patient was previously treated elsewhere with multiple topical therapies, without response. These included the class IV steroid triamcinolone acetonide 0.1% and the class II steroid fluocinonide, as well as calcipotriene ointment.

Physical Examination

- Physical examination revealed diffuse erythroderma with “islands of sparing” and follicular prominence, facial and scalp erythema and scaling, as well as marked hyperkeratosis of the palms and soles.
- His affected body surface area (BSA) exceeded 80%.
- The patient reported symptoms of pruritus as well as painful hands and feet, which severely limited his ability to work.
- Biopsy was consistent with a diagnosis of PRP.

Initial Treatments and Response

- The patient was treated with the class II topical steroid halobetasol and UVB light therapy for a 3-month period.
 - The erythroderma improved, but the patient’s hands and feet failed to respond.
- The patient was then lost to follow-up for 18 months.
- When he returned to the clinic, the hyperkeratosis and fissuring of his hands and feet were incapacitating; however, his disease remained controlled over other areas of his body.
- The patient received acitretin for 4 months, but this treatment was ineffective.

Efalizumab Therapy

Treatment

- In December 2006, efalizumab subcutaneous monotherapy was initiated at a conditioning dose of 0.7 mg/kg, followed by treatment with 1 mg/kg/wk thereafter.

Response

- Within 3 weeks, the patient experienced 75% improvement of his PRP, with marked diminution of hyperkeratosis and a return of function (Figure 1).
- Efalizumab was discontinued after 3 doses because the patient was unable to pay for subsequent therapy, but it was reinstated 6 weeks later.
 - During the hiatus, a slight flare was noted.
- Independent of the attending physician’s counsel, the patient has pursued treatment every other week.

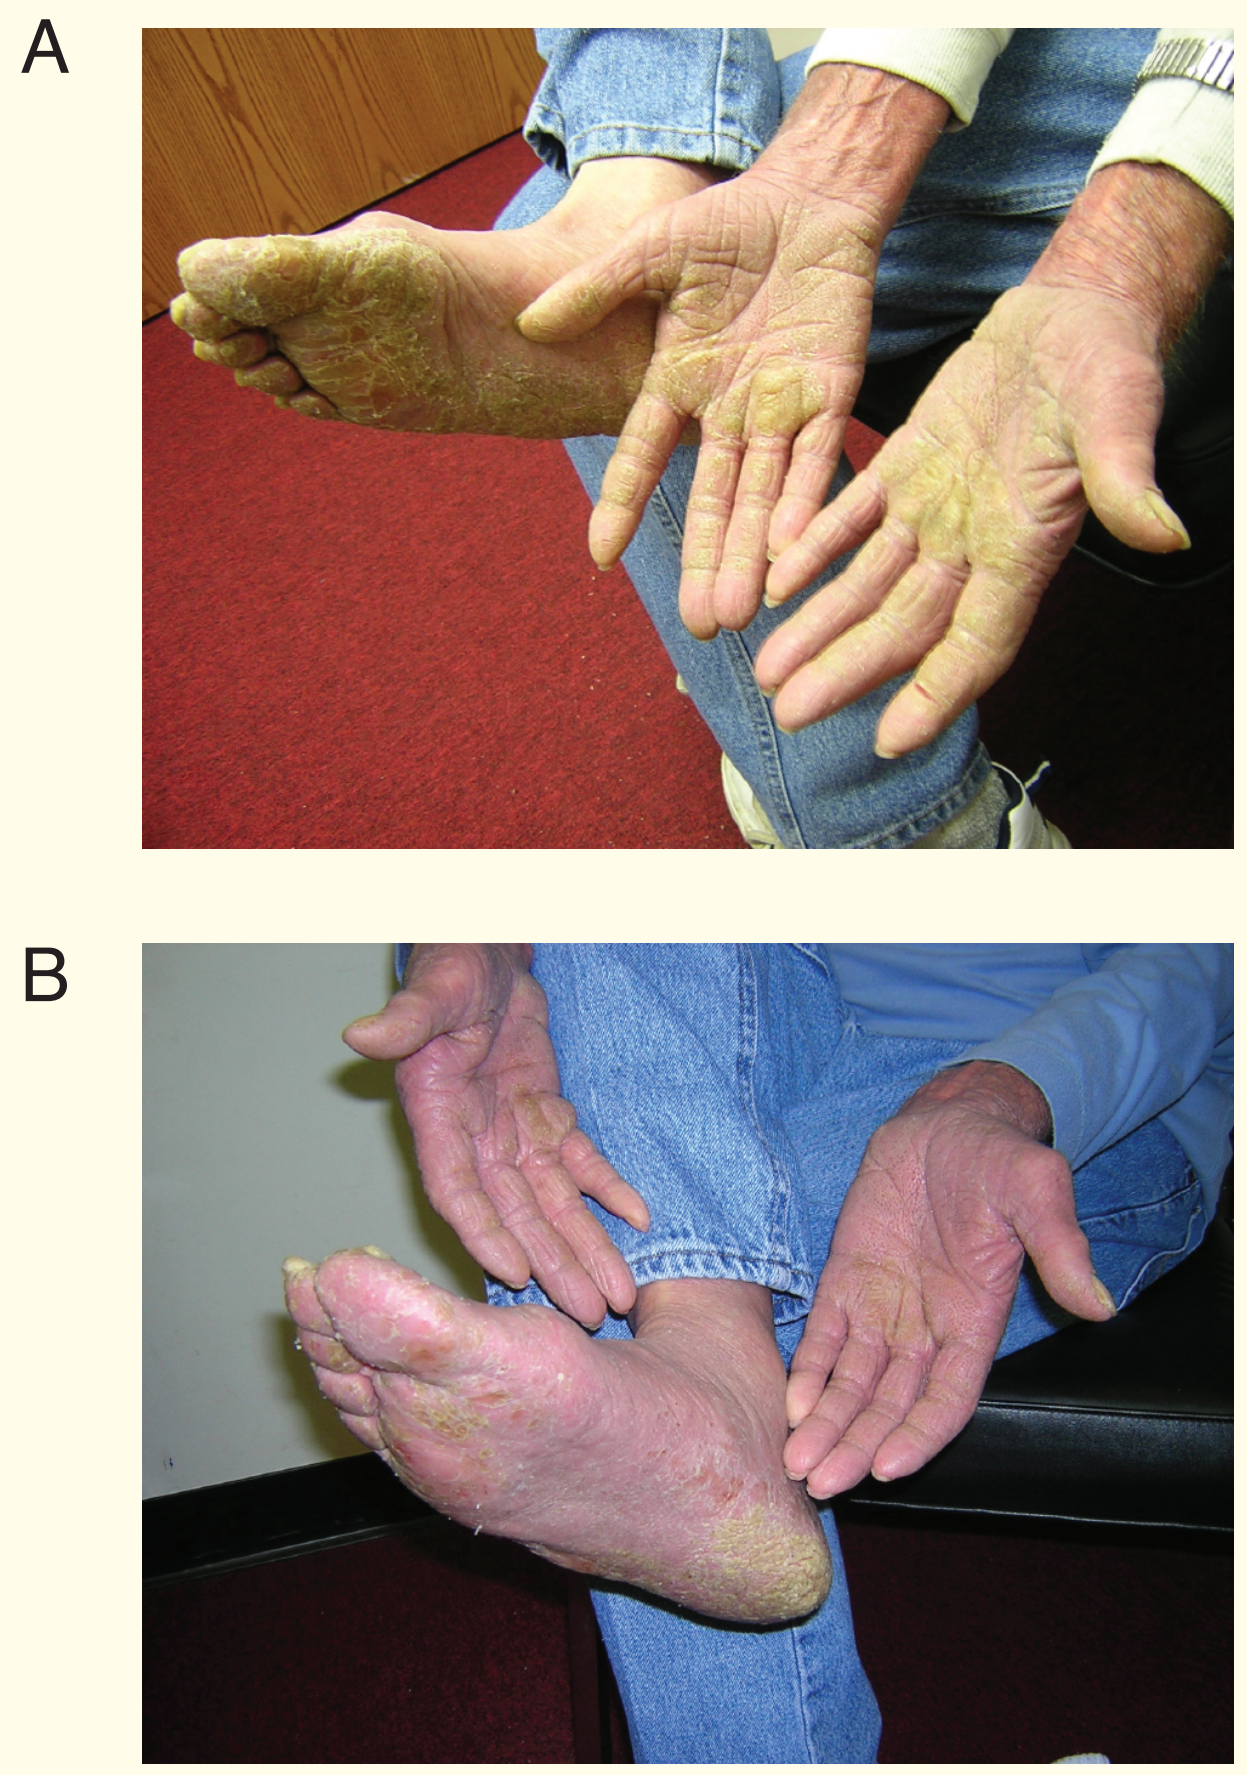
A
B

Figure 1. A: Before treatment with efalizumab. B: After 3 weeks’ efalizumab treatment.

Follow-up

- The patient has continued treatment with efalizumab injections every other week for more than 3 months and has not experienced any adverse events.
- His condition has remained stable.

CONCLUSIONS

- A patient with PRP was successfully treated with efalizumab.
- The response of this patient to efalizumab suggests T-cell involvement in the pathogenesis of PRP.
- Efalizumab may be useful in the management of patients with recalcitrant PRP.

REFERENCES

1. Vanderhooft SL, et al. *Arch Dermatol.* 1995;131:448-453.
2. Wells RS; in discussion on Borrie P. *Br J Dermatol.* 1967;79:115-116.
3. Griffiths WAD. *Clin Exp Dermatol.* 1976;1:37-50.
4. Griffiths WAD. *Clin Exp Dermatol.* 1980;5:105-112.
5. Albert MR, Mackool BT. *Int J Dermatol.* 1999;38:1-11.
6. Griffiths A. *J Am Acad Dermatol.* 1984;10:1086-1088.
7. Miralles ES, et al. *Br J Dermatol.* 1995;133:990-993.
8. Judge MR, et al. In: Burns T, et al, eds. *Rook’s Textbook of Dermatology.* Vol 2. 7th ed. Oxford, UK: Blackwell Science; 2004:34.1-111.
9. Goldsmith L, Baden H. In: Freedberg IM, et al, eds. *Fitzpatrick’s Dermatology in General Medicine.* Vol 1. 6th ed. New York: McGraw-Hill; 2003:442-444.
10. Wetzig T, Sticherling M. *Br J Dermatol.* 2003;149:202-203.
11. Coras B, et al. *Br J Dermatol.* 2005;152:388-389.
12. Manoharan S, et al. *Australas J Dermatol.* 2006;47:124-129.
13. Huber A, et al. *Acta Derm Venereol.* 2006;86:254-255.
14. Weinberg JM, Siegfried EC. *Arch Dermatol.* 2006;142:555-558.
15. Kaelin U, et al. *J Am Acad Dermatol.* 2006;55:529-532.
16. Cheng A, Mann C. *Arch Dermatol.* 2006;142:680-682.
17. Gómez M, et al. *J Drugs Dermatol.* 2007;6:337-339.
18. Lotsikas-Baggili AJ, et al. Presented at: American Academy of Dermatology Annual Meeting; February 2-6, 2007; Washington, DC. Poster 2762.
19. Klein A, et al. *Dermatology.* 2007;215:72-75.

The development of this poster was sponsored by Genentech, Inc.

Efalizumab is approved for use in patients with chronic moderate to severe plaque psoriasis; the use of efalizumab to treat pityriasis rubra pilaris is considered to be “off-label.”